Synthesis and antiproliferative activity of novel amino substituted tetracyclic imidazo[4,5-*b*]pyridine derivatives

M. Hranjec[1], N. Perin[1], I. Boček[1], B. Lučić[2], M. Kralj[3] and M. Mioč[3]

[1] Department of Organic Chemistry, Faculty of Chemical Engineering and Technology University of Zagreb, Marulićev trg 20, HR-10000 Zagreb, Croatia; [2] Pliva d.o.o, TAPI I&R, 10000 Zagreb, Savski Marof, Croatia; [3] Institute Rudjer Bošković, Laboratory of Experimental Therapy, Bijenička cesta 54, 10000 Zagreb, Croatia

Due to the structural similarity of imidazo-pyridine heterocyclic system with naturally occurring purines and great therapeutic potential and significance, suchlike derivatives nowadays play an important role in medicinal chemistry and drug discovery. Imidazo-pyridine derivatives foregrounded their importance in the prevention of proper functioning of cancerous cells, diseases related to the central nervous system, inflammation, *etc*. Taking into account a great biological potential and the fact that imidazo[4,5-*b*]pyridine scaffold is among the most privileged and important building blocks in medicinal chemistry as well as our previously published significant biological results of imidazo[4,5-*b*]pyridine derivatives, we have designed and synthesized novel tetracyclic derivatives as novel and potent antiproliferative agents. The synthesis of all newly prepared compounds was conducted using conventional methods of organic synthesis and microwave assisted synthesis. Tetracyclic derivatives were substituted with chosen amino side chains which have significantly enhanced the antiproliferative activity of tetracyclic derivatives and are placed at the different position of skeleton. Additionally, the impact of the N atom position in pyridine nuclei on biological activity was studied. The antiproliferative activity of regioisomers was studied against human cancer cells and non-tumour cells. As a standard drug *etoposide* was used and interestingly, the majority of compounds showed improvement of antiproliferative activity on HCT116 and MCF-7 cancer cells when compared to *etoposide*. From the obtained results it could be noticed that the position of N nitrogen in pyridine ring has strong impact on the antiproliferative activity while the type of amino substituent did not influence activity signifcantly. Thus, regioisomers 6, 7 and 9 substituted with amino substistuents at position 2 showed noticeable enhancement of activity in comparison to their counterparts 10, 11 and 13 having IC_{50} values from 0.3 mM to 0.9 mM against all three cancer cells.

Table 1. Antiproliferative activity of tested compounds and their clogP values

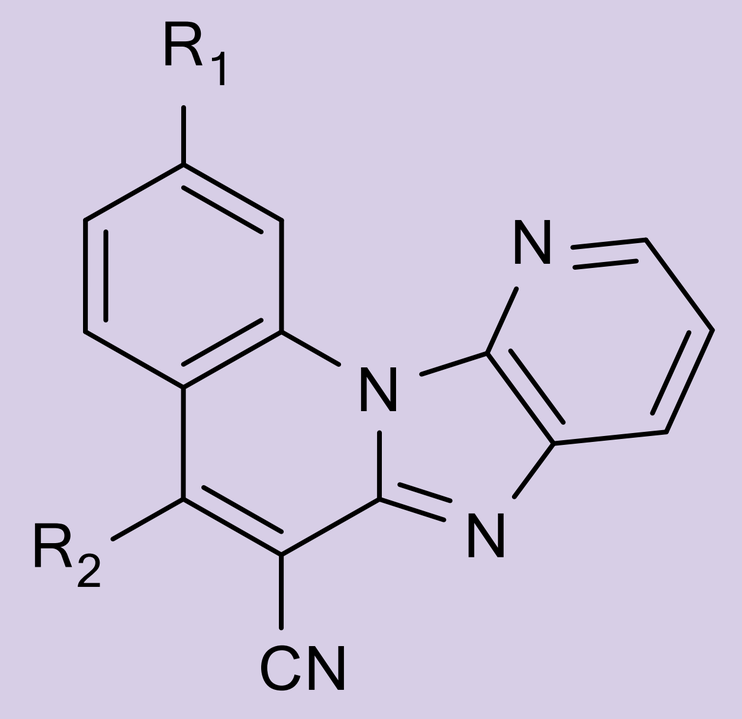
R1
R2
N
N
N
CN

5a, 6-9, 20-21, 24-30

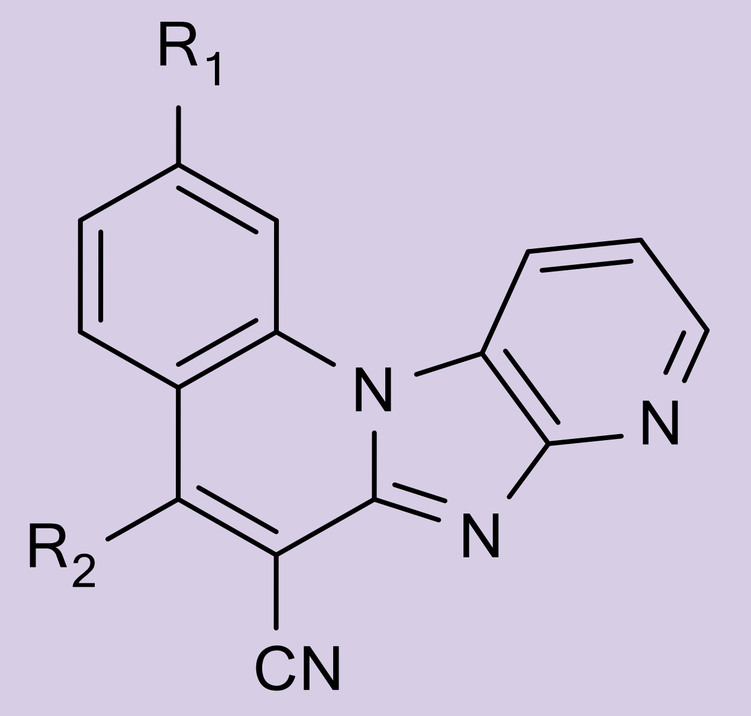
R1
R2
N
N
N
CN

5b, 10-13, 22-23, 31-37

Compd	R_1	R_2	IC_{50}[a] (μM) *HCT116*	*MCF-7*	*H460*	*HEK 293*	clogP[b]
5a	F	H	5±1.4	≥100	≥100	n.t.[c]	2.86
5b	F	H	≥100	≥100	≥100	n.t.	2.86
6	$NH(CH_2)_3N(CH_3)_2$	H	0.4±0.08	0.7±0.1	0.4±0.1	0.3±0.08	3.34
7	$NHCH_2CH(CH_3)_2$	H	0.5±0.02	0.4	0.3±0.05	0.5±0.2	4.26
8	piperidine	H	5.5±1.5	3±0.9	≥100	n.t.	3.87
9	piperazine	H	0.4±0.06	0.4±0.1	0.4±0.08	0.7±0.4	2.47
10	$NH(CH_2)_3N(CH_3)_2$	H	2.3±0.5	12±4.5	11±1.7	4±0.09	3.34
11	$NHCH_2CH(CH_3)_2$	H	3±1.7	3±2	12±4	n.t.	4.26
12	piperidine	H	10±2.7	5±4.5	49±0.01	n.t.	3.87
13	piperazine	H	0.9±0.8	3.1±1.2	7±0.7	n.t.	2.47
20	H	Cl	3±3	6±4	≥100	n.t.	3.30
21	F	Cl	3±0.06	2 ±0.6	≥100	n.t.	3.44
22	H	Cl	0.1±2	2±0.4	4.6±1.6	1.6±0.1	3.30
23	F	Cl	3.5±2	2±0.8	≥100	n.t.	3.44
24	H	$NH(CH_2)_3N(CH_3)_2$	0.9±0.03	2.9±1.3	1.4±0.2	1.8±0.2	3.72
25	H	$NHCH_2CH(CH_3)_2$	≥100	≥100	≥100	>100	4.64
26	H	piperidine	47±0.6	16±1.3	≥100	n.t.	4.08
27	H	piperazine	3.6±0.3	4.7±0.5	1.8±1	n.t.	2.68
28	$NHCH_2CH(CH_3)_2$	$NHCH_2CH(CH_3)_2$	3±0.2	1.8±0.3	5.4±0.4	n.t.	5.78
29	piperidine	piperidine	1±0.8	0.5±0.2	≥100	>100	5.04
30	piperazine	piperazine	0.4±0.05	0.4±0.0	0.7±0.4	0.2±0.1	2.25
31	H	$NH(CH_2)_3N(CH_3)_2$	0.8±0.1	3.2±0.5	2±0.1	2.6±0.7	3.72
32	H	$NHCH_2CH(CH_3)_2$	14±2.7	14±3.5	17.5±0.7	>100	4.64
33	H	piperidine	≥100	≥100	≥100	n.t.	4.08
34	H	piperazine	3.5±1.2	3±2.8	7±0.6	n.t.	2.68
35	$NHCH_2CH(CH_3)_2$	$NHCH_2CH(CH_3)_2$	3.7±0.4	2 ±0.7	7.6±0.5	n.t.	5.78
36	piperidine	piperidine	1.8±0.1	3.6±0.6	7±3.5	n.t.	5.04
37	piperazine	piperazine	≥100	≥100	≥100	>100	2.25
Etoposide	-	-	5±2	1±0.7	0.1±0.04	-	-

[a] IC_{50}; the concentration that causes 50% growth inhibition; [b] clogP; calculated logP values were obtained by using ChemDraw Professional 15.0.; c) not tested.

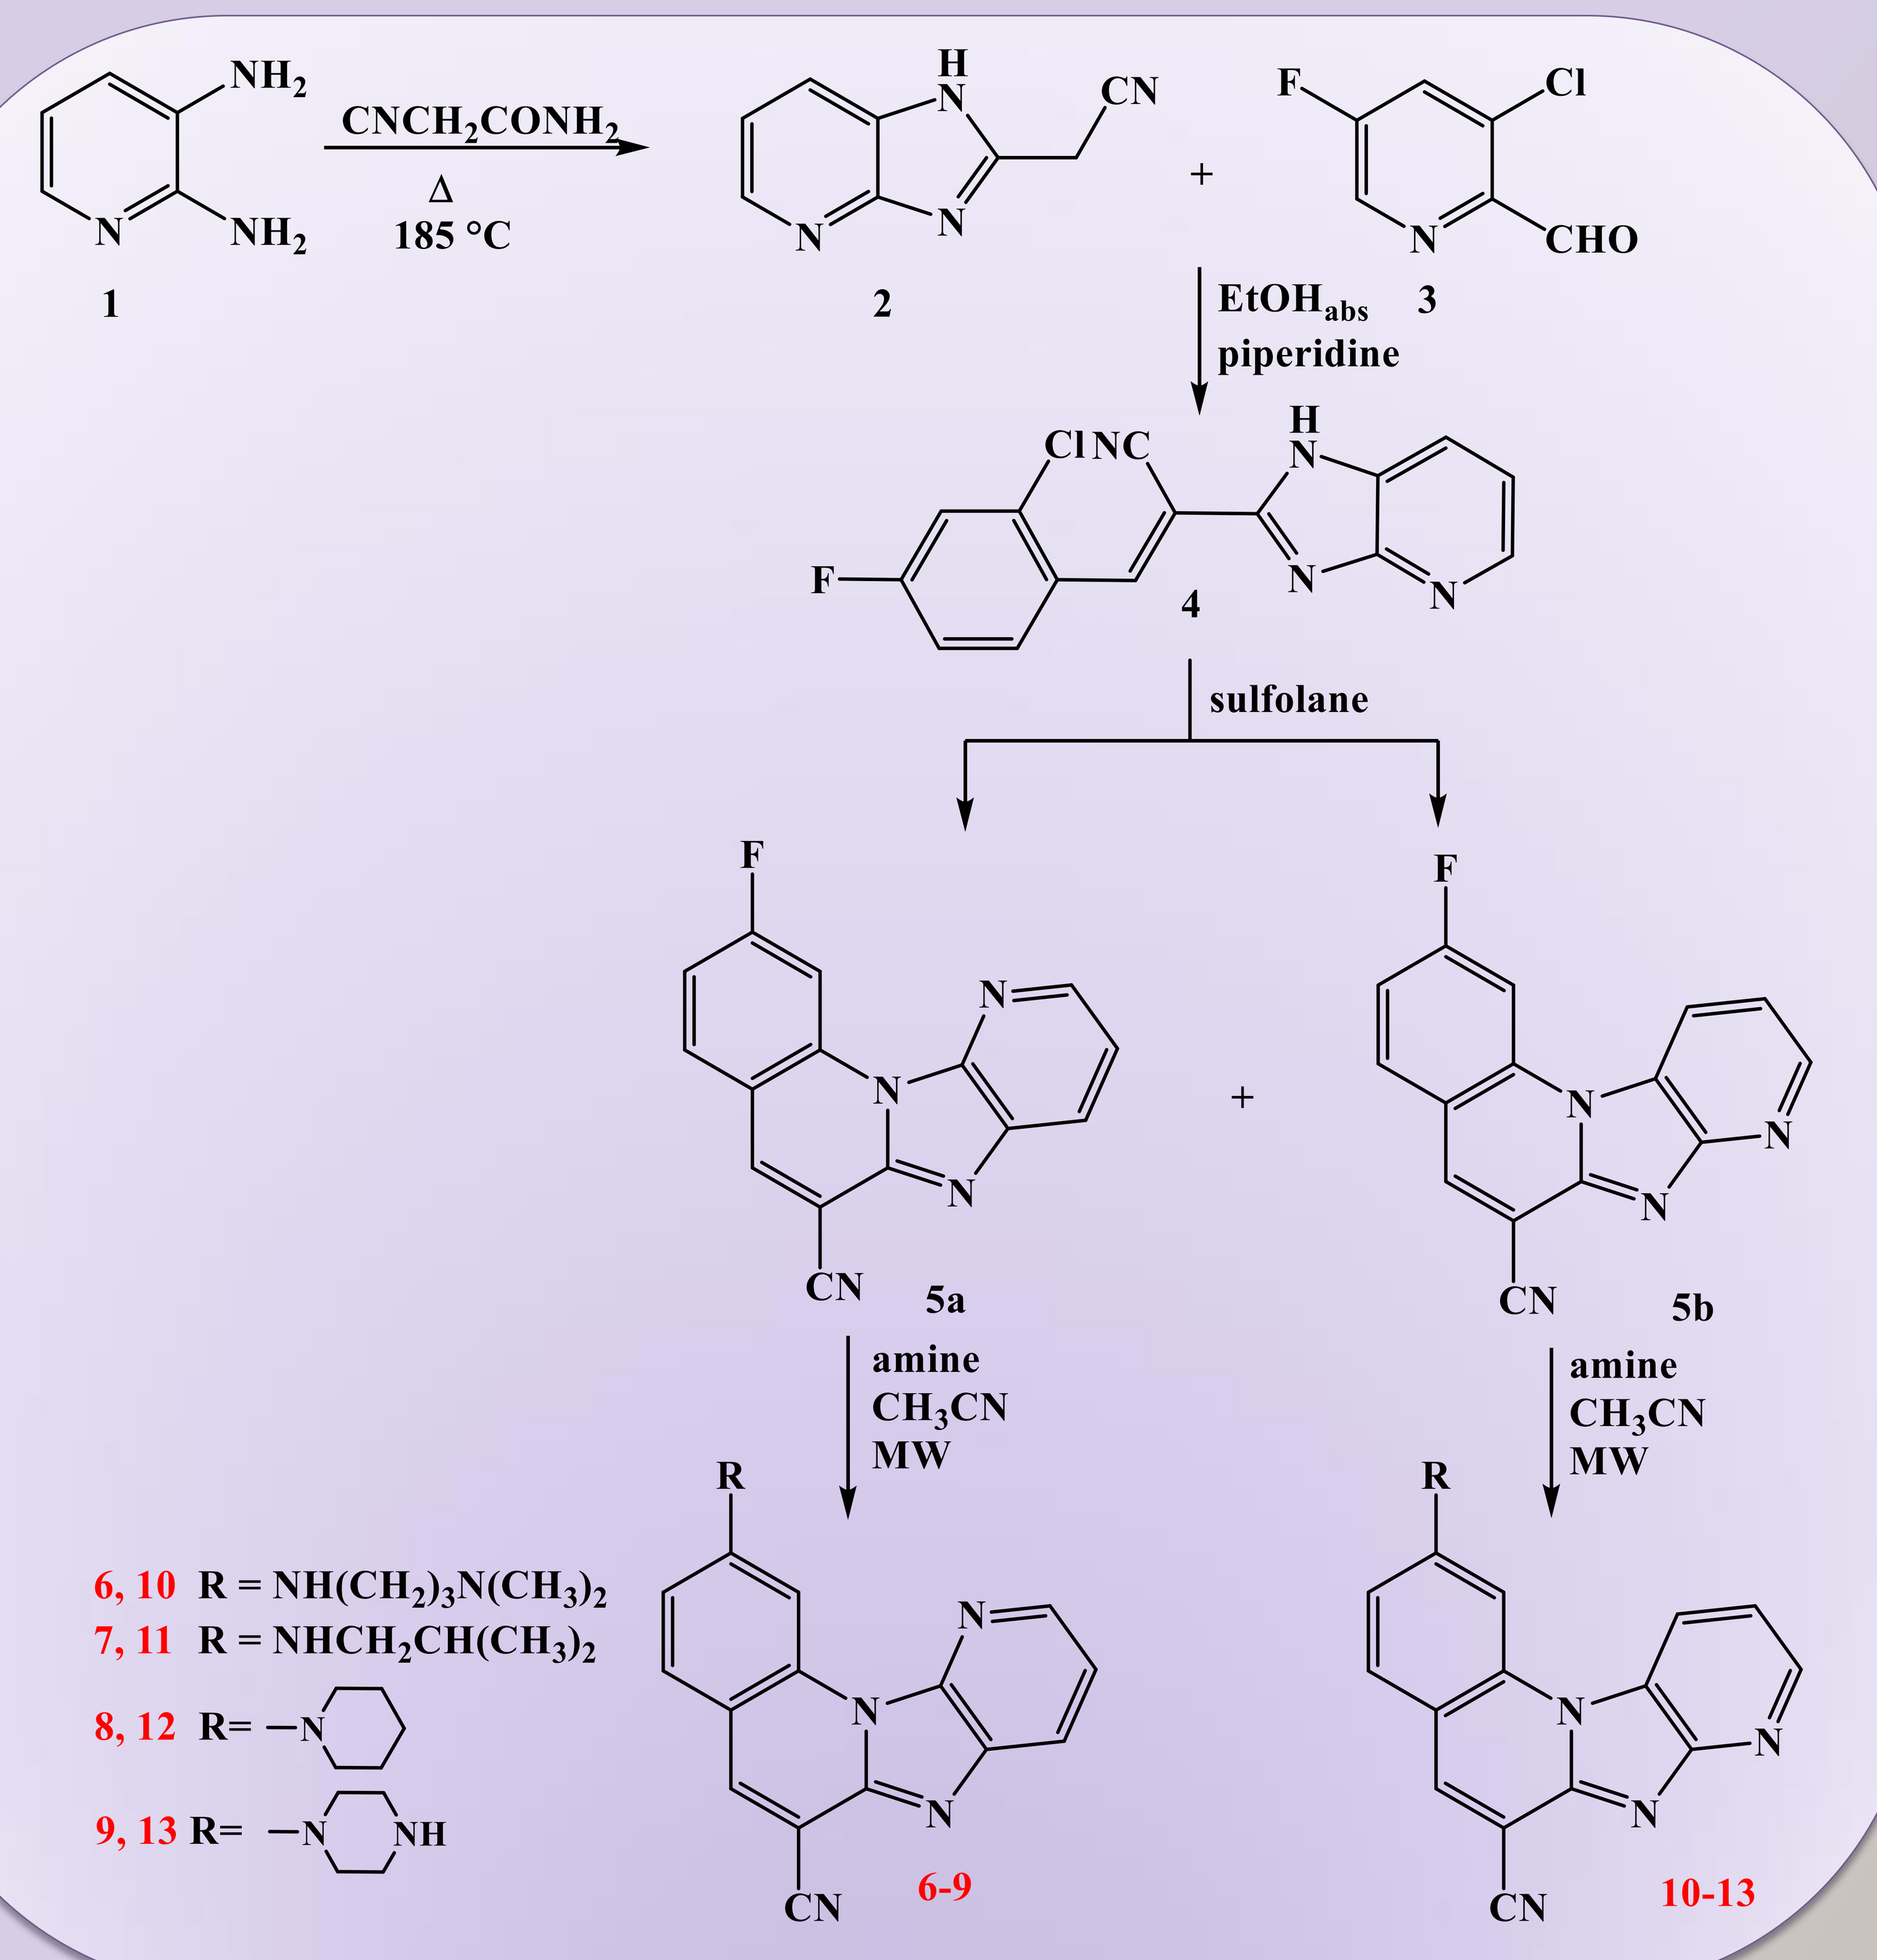
CNCH2CONH2
Δ
185 °C
1
2
3
EtOHabs
piperidine
4
sulfolane
5a
5b
amine
CH3CN
MW
6, 10 R = NH(CH2)3N(CH3)2
7, 11 R = NHCH2CH(CH3)2
8, 12 R=
9, 13 R=
6-9
10-13

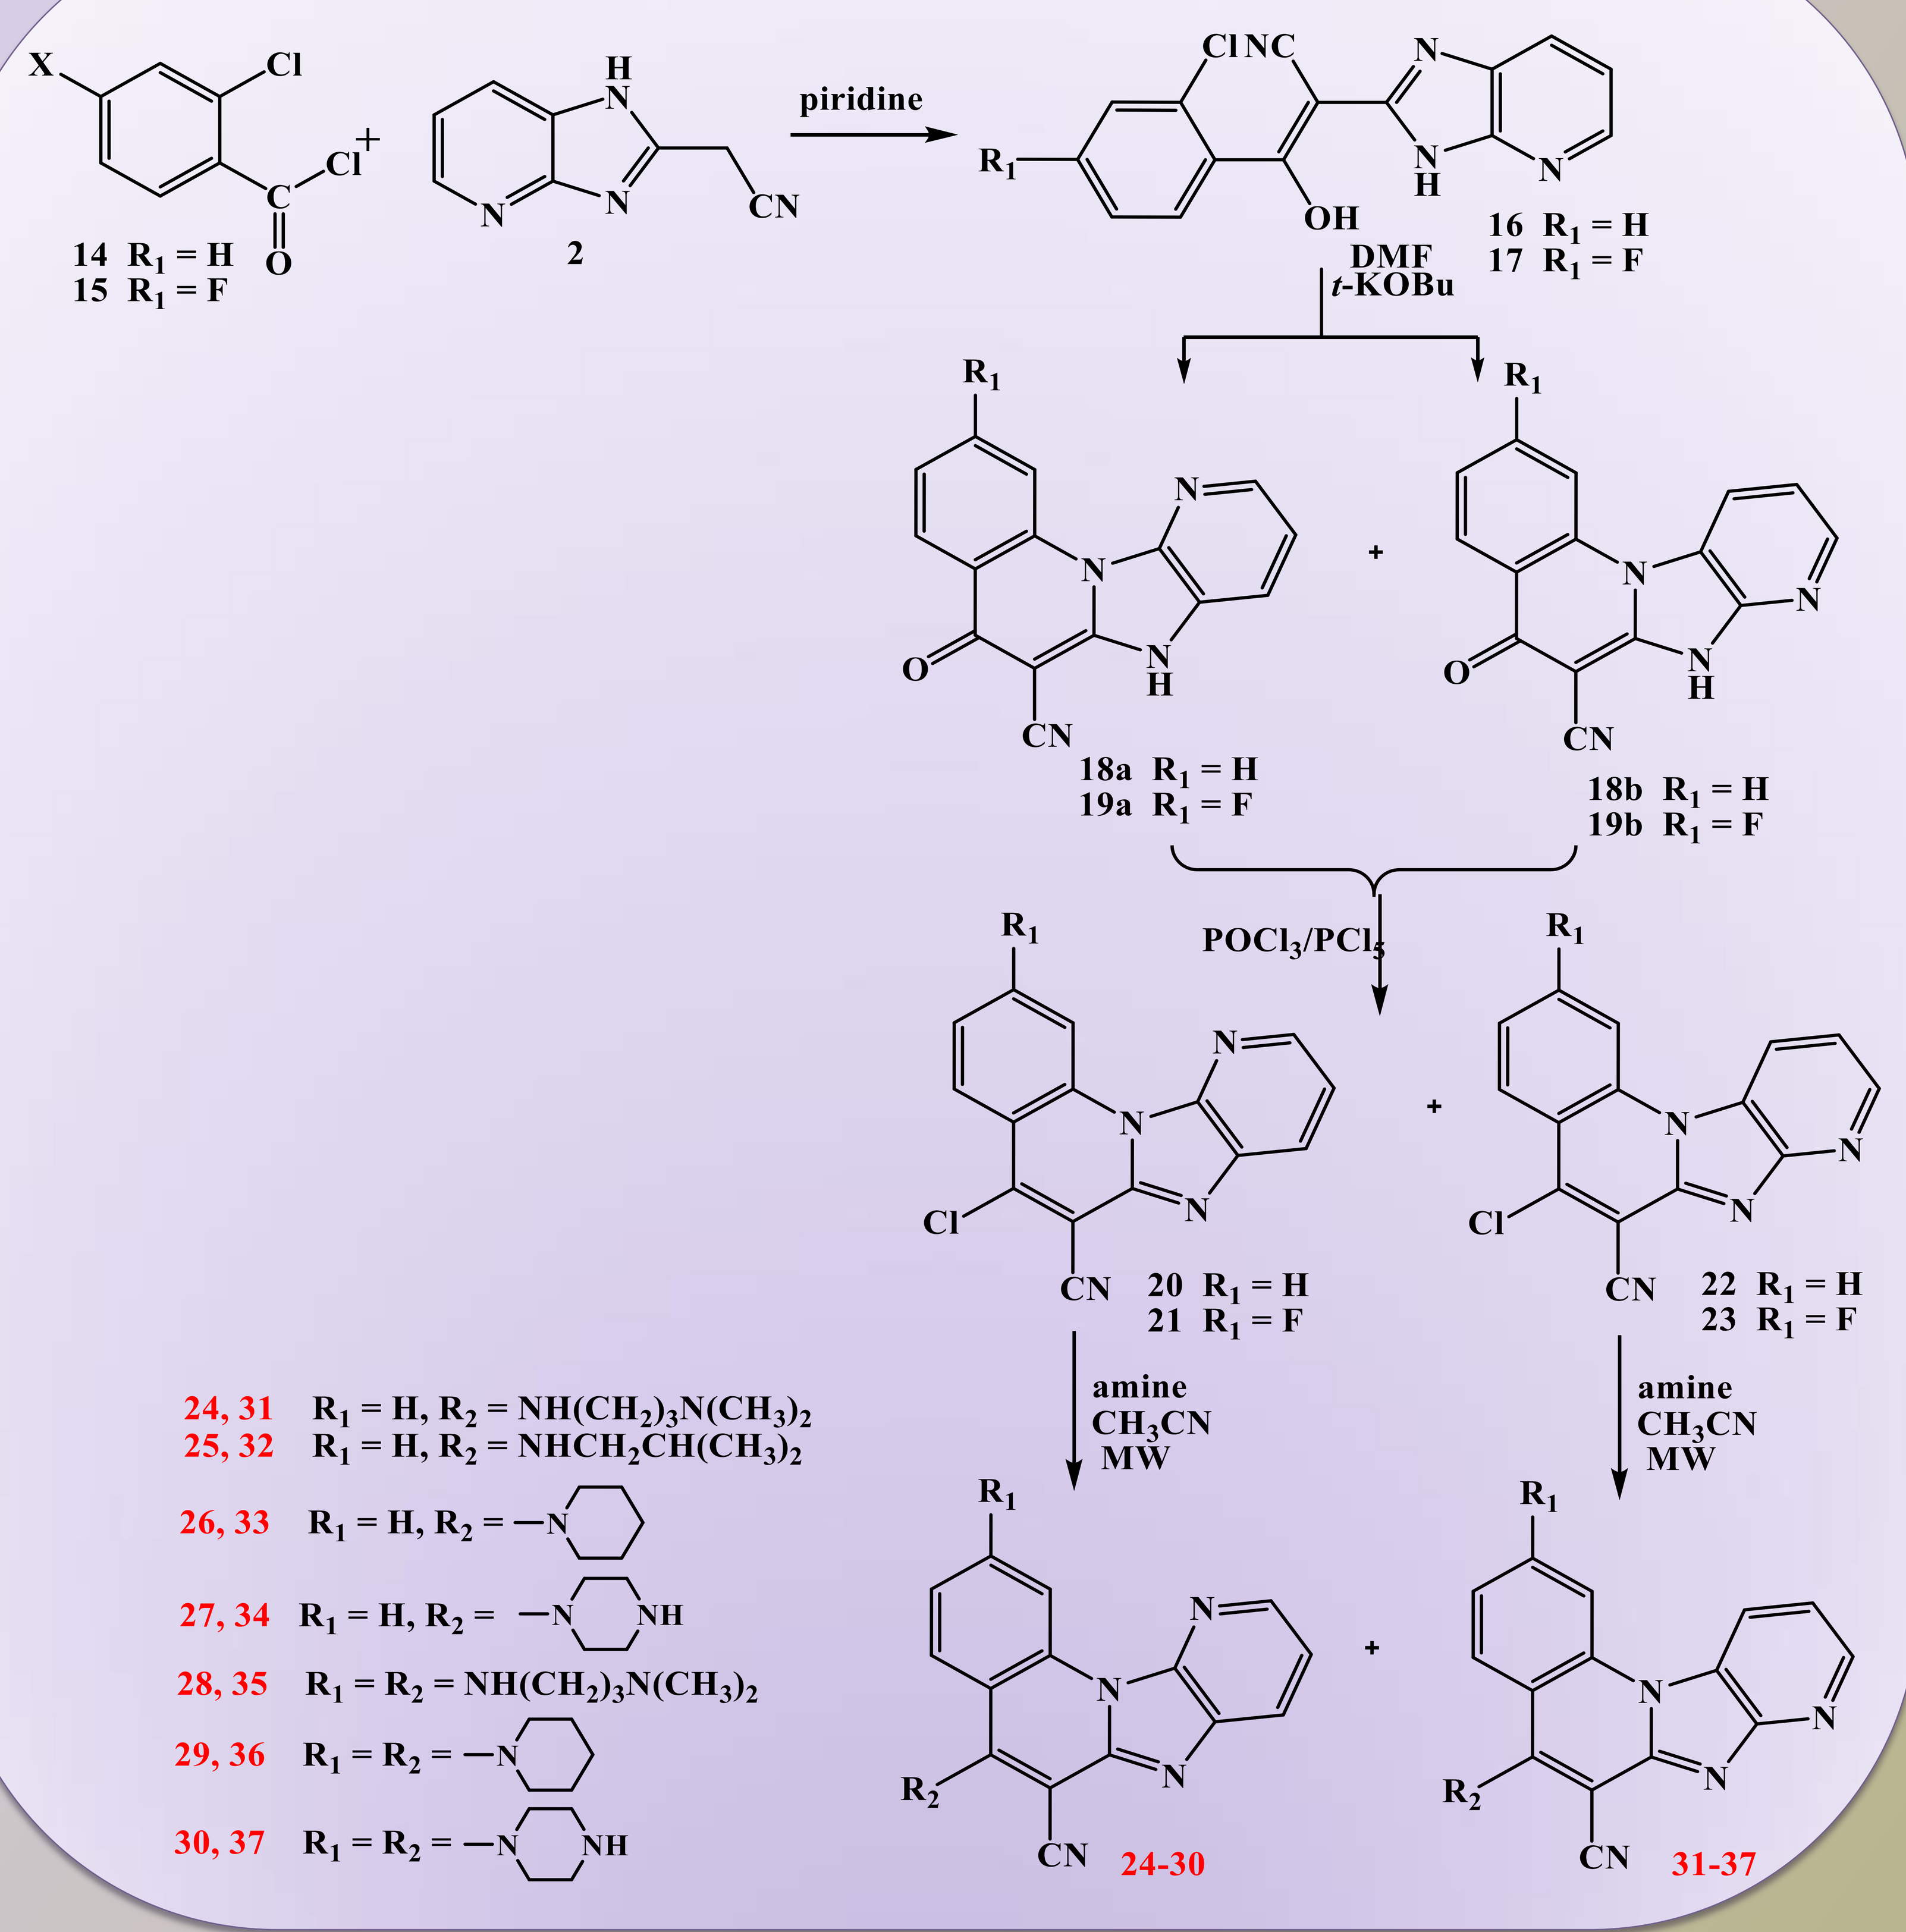
14 R1 = H
15 R1 = F
2
piridine
16 R1 = H
17 R1 = F
DMF
t-KOBu
18a R1 = H
19a R1 = F
18b R1 = H
19b R1 = F
POCl3/PCl5
20 R1 = H
21 R1 = F
22 R1 = H
23 R1 = F
amine
CH3CN
MW
24, 31 R1 = H, R2 = NH(CH2)3N(CH3)2
25, 32 R1 = H, R2 = NHCH2CH(CH3)2
26, 33 R1 = H, R2 =
27, 34 R1 = H, R2 =
28, 35 R1 = R2 = NH(CH2)3N(CH3)2
29, 36 R1 = R2 =
30, 37 R1 = R2 =
24-30
31-37

1. Hranjec M, Lučić B, Ratkaj I, Pavelic SK, Piantanida I, Pavelic K, Karminski-Zamola G (2011) Novel imidazo[4,5-b]pyridine and triaza-benzo[c]fluorene derivatives: Synthesis, antiproliferative activity and DNA binding studies. Eur J Med Chem 46:2748–2758.
2. Newhouse BJ, Wenglowsky S, Grina J, Laird ER, Voegtli WC, Ren L, Ahrendt K, Buckmelter A, Gloor SL, Klopfenstein N, Rudolph J, Wen Z, Li X, Feng B (2013) Imidazo[4,5-b]pyridine inhibitors of B-Raf kinase, Bioorg Med Chem Lett 23:5896-5899.
3. Perin N, Nhili R, Ester K, Laine W, Karminski-Zamola G, Kralj M, David-Cordonnier MH, Hranjec M (2014) Synthesis, antiproliferative activity and DNA binding properties of novel 5-Aminobenzimidazo[1,2-*a*]quinoline-6-carbonitriles. Eur J Med Chem 80:218–227.
4. Perin N, Martin-Kleiner I, Nhili R, Laine W, David-Cordonnier MH, Vugrek O, Karminski-Zamola G, Kralj M, Hranjec M, Biological activity and DNA binding studies of 2-substituted benzimidazo[1,2-*a*]quinolines bearing different amino side chains, Med Chem Comm 4 :1537–1550.

This work was funded by the Croatian Science Foundation under the project 4379, AntioxPot.